A novel nucleic acid simplification procedure capable of detecting multiple strains of both DNA and RNA viruses in a single reaction

Nicky Boulter, Shoo Peng Siah, Neralie Coulston, Cassandra Vockler, Kristina Warton, Pooli Rajasekariah, John Melki and Douglas Millar

Introduction:

A rapid and sensitive assay that is able to specifically detect a range of clinically important DNA and RNA viruses from a single sample would be of enormous benefit to the healthcare sector. Molecular diagnostic assays are of particular importance when the infectious agent is unable to be grown in conventional culture, such as for Human Papilloma Virus (HPV). However, the design of these molecular assays can be technically challenging as many pathogens contain diverse subtypes that can all be involved in diseases, such as in the many genotypes or serotypes of HPV, HCV and enteroviruses. We have developed an assay that successfully and specifically detects a wide range of DNA and RNA viruses. The viral discrimination is achieved through the conversion of the viral nucleic acid with sodium bisulphite.

Treatment of DNA with sodium bisulphite results in the chemical conversion of all unmethylated cytosines to thymine via a uracil intermediate, effectively converting a 4 base genome into a 3 base genome. This technology can be utilised to reduce the complexity of the genome of different organisms or strains so that they are more similar to each other, thus facilitating the detection of multiple organisms/strains in a single PCR reaction without the need for multiplexing. However, treatment of RNA with sodium bisulphite under the same conditions results in complete degradation of the RNA and is of no clinical utility. We have developed a new method for the simplification of RNA using sodium bisulphite. An example of how simplification can be applied to the detection of different HCV strains is shown in Table 1.

Detection of different HCV strains by DNA simplification

HCV Strain	Before Simplification	After Simplification
1a	CAAGTTCCCGGGTGGCGG	TAAGTTTTTGGGTGGTGG
1b	CAAGTTCCCGGGCGGTGG	TAAGTTTTTGGGTGGTGG
1c	TAAGTTCCCGGGTGGCGG	TAAGTTTTTGGGTGGTGG
2a	TAAGTTTCCGGGCGGCGG	TAAGTTTTTGGGTGGTGG
2b	CAAGTTCCCGGGTGGCGG	TAAGTTTTTGGGTGGTGG
2c	TAAGTTCCCGGGCGGTGG	TAAGTTTTTGGGTGGTGG
2k	CAAGTTCCCGGGCGGTGG	TAAGTTTTTGGGTGGTGG
3a	TAAGTTCCCGGGTGGCGG	TAAGTTTTTGGGTGGTGG
3b	TAAGTTCCCGGCTGGCGG	TAAGTTTTTGGTTGGTGG
3k	TAAGTTCCCAGGCGGCGG	TAAGTTTTTAGGTGGTGG
4a	TAAGTTCCCGGGTGGTGG	TAAGTTTTTGGGTGGTGG
5a	CAAGTTCCCGGGCGGTGG	TAAGTTTTTGGGTGGTGG
6a	CAAGTTCCCGGGTGGCGG	TAAGTTTTTGGGTGGTGG
6b	CAAGTTCCCGGGCGGCGG	TAAGTTTTTGGGTGGTGG
6g	CAAGTTCCCGGGCGGTGG	TAAGTTTTTGGGTGGTGG
6h	CAAGTTCCCGGGCGGCGG	TAAGTTTTTGGGTGGTGG
6k	TAAGTTCCCGGGTGGCGG	TAAGTTTTTGGGTGGTGG
Consensus	YAAGTTYCCRGSYGGYGG	TAAGTTTTTRGKTGGTGG
	66% homology over 18 bases 64 possible primer combinations	89% homology over 18 bases 4 possible primer combinations

Table 1: Consensus sequence of HCV genotypes before and after RNA simplification. The simplification has resulted in an increased homology from 66% to 89%.

To demonstrate the performance of our assay, we have validated it using a series of commercially available HCV performance, linearity, genotyping panels and finally on a blinded panel of 138 clinical samples.

We have also developed a single tube broad-range assay that successfully detects all High Risk HPV (HR-HPV) sub-types (16, 18, 31, 33, 35, 39, 45, 51, 52, 56, 58, 59 and 68). The assay was used during a clinical trial that compared the sensitivity (false negative) and selectivity (false positive) rates of the hcII HPV DNA test (Digene Corp., USA) against our HR HPV DNA Test using PreservCyt Solution (Cytyc Corp., Marlborough USA) specimens obtained from the ThinPrep® Pap Test™.

Furthermore, we have developed prototype assays to detect a range of additional DNA and RNA viruses, including HBV, HSV-1, HSV-2, CMV, EBV, HIV-1 and EV.

Methods:

HPV assay and clinical trial: The clinical trial was performed by the South Eastern Area Laboratory Services (SEALS) of the Prince of Wales Hospital, Randwick, NSW, Australia (Principal Investigator: Prof. William Rawlinson). The study design was a double blind, controlled, crossover, comparative study of the sensitivity (false negative) and selectivity (false positive) rates of the hcII Human Papillomavirus DNA Test (Digene Corporation, USA) versus the Human Genetic Signatures HR-HPV DNA Test (as compared to the MY09/MY11 and the GP5+/GP6+ L1 PCR based "Gold Standard PCR Test" involving genotyping via sequencing). Both tests were performed exactly as per the recommendations of the manufacturer or according to the literature. The primary endpoint of the study was the Positive Predictive Value of each test in determining the presence of High Risk HPV in a sample. Sensitivity, Specificity and Negative Predictive Value were secondary endpoints. A total of 834 samples were analysed during the trial.

HCV assay: HCV RNA samples were obtained from Acrometrix (OptiQual HCV high positive control), or BBI diagnostics (HCV RNA linearity panel PHW804, and Worldwide HCV genotype panel WWHV302) and purified with the QiaAmp Ultrasens Viral purification kit according to the manufacturer's instructions (Qiagen). Blinded clinical samples were obtained from a local hospital and purified as above. Samples were treated with sodium bisulphite, desulphonated and 11µl of the converted HCV RNA samples were reverse transcribed with Superscript III reverse transcriptase (Invitrogen) or iScript reverse transcriptase (Biorad) using modified random primers. One tenth of the cDNA was then subjected to PCR amplification with primers specific for converted HCV RNA. PCR was performed in 50µl reactions using 1.5X Promega master mix and 100ng each of forward and reverse primers and cycled at 95°C, 3 mins; [95°C, 10 secs; 53°C, 1 min; 68°C, 1 min] x40 in a Hybaid PX2 thermal cycler.

DNA viruses: Epstein Barr virus (EBV), Hepatitis B virus (HBV), Human simplex virus-1 and -2 (HSV-1, HSV-2) and Cytomegalovirus (CMV) positive controls were purchased from Acrometrix (Acrometrix Corporation, CA, USA). DNA was purified using the QiaAmp Ultrasens viral extraction kit as per the manufacturer's instructions (Qiagen). The DNA was diluted to represent a range of viral concentrations and the samples were then bisulphite modified using the Human Genetic Signatures MethylEasy Xceed™ kit (Sydney, Australia) as per the manufacturer's instructions, with the exception that the bisulphite was removed by isopropanol precipitation instead of column purification. 2µl of each sample was amplified using viral-specific primers in a one round or nested PCR reaction. First and second round PCRs were performed in 50µl reactions as above using 30 cycles of PCR with annealing temperatures between 55-60°C. 20µl of each sample was electrophoresed on a 2% precast agarose e-gel (Invitrogen).

RNA viruses: Human Immunodeficiency Virus (HIV-1) and Enterovirus (EV) positive controls were purchased from Acrometrix (Acrometrix Corporation, CA, USA). RNA was purified, bisulphite converted and reverse transcribed as described for HCV. Nested PCR amplification and gel analysis was carried out as already stated using 40 cycles with annealing temperatures of 50-55°C.

Results:

Clinical Trial: The SEALS clinical trial results demonstrated that the HGS HR-HPV assay had a statistically significant higher positive predictive value than the Digene test ($P < 0.001$; Table 2) and thus the false positive detection rate for the HGS test is lower than that of the Digene test (19.9% vs 28.4%; $P < 0.001$). The negative predictive value of the HGS test and the Digene test was 80.4% and 80.1%, respectively, but this was not statistically significant ($P=0.677$).

	Reference method (PCR)		Sensitivity (%)	Specificity (%)	PPV (%)	NPV (%)
	+	-				
HG test results						
+	197	49				
-	115	473	63.1	90.6*	80.1*	80.4
Digene HcII test results						
+	202	80				
-	110	442	64.7	84.6	71.6	80.1

Table 2: HGS and Digene test results compared with the Reference Standard method (PCR with MY and GP primer sets) for 834 cervical specimens. PPV = positive predictive value; NPV = negative predictive value; *p<0.001 by two-tailed student's *t*-test comparing result with HGS and Digene hcII tests for detection of HPV in cervical samples. C. Baleriola, D. Millar et al, J. Clin. Microbiol. (2008)

HCV assay: An example of the results obtained when using our RNA bisulphite conversion RT-PCR assay is shown in Figure 1. The results of the linearity panel and dynamic range demonstrate that there is good sensitivity for the detection of HCV viral RNA, using our simplification method, ranging from 1 562 500 IU down to 23 IU, illustrating that the assay can detect viral RNA over a very broad range of concentrations.

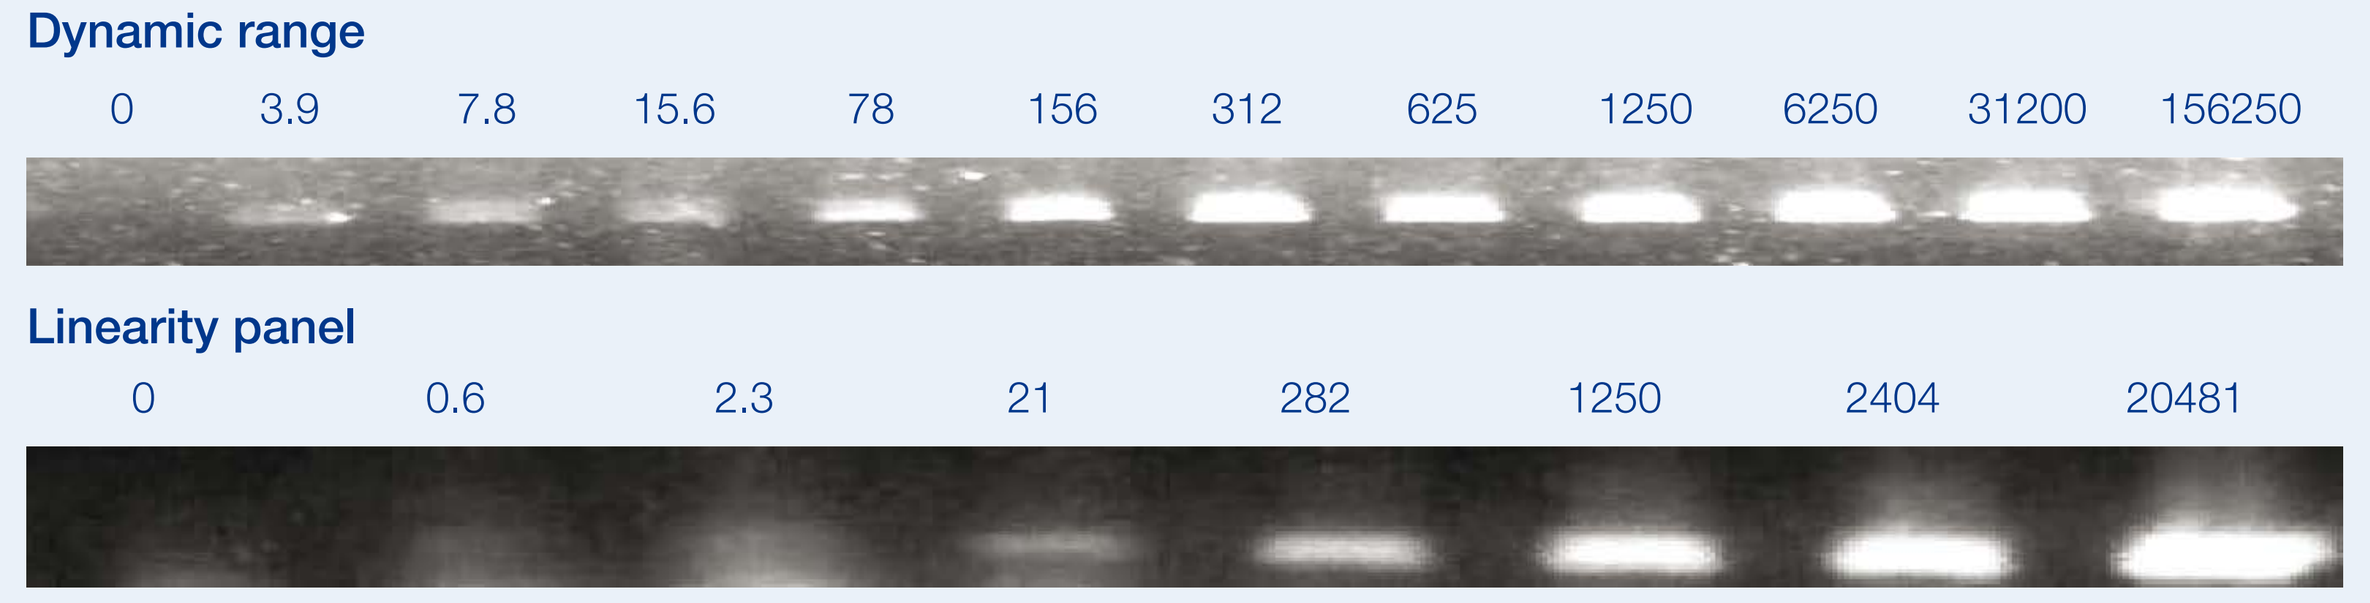
Dynamic range
0 3.9 7.8 15.6 78 156 312 625 1250 6250 31200 156250
Linearity panel
0 0.6 2.3 21 282 1250 2404 20481

Figure 1: A HCV linearity panel from BBI diagnostics or a dynamic range of HCV RNA concentrations from 39 IU to 1 562 500 IU/mL were purified from Acrometrix Optiqual HCV high positive control, bisulphite converted, reverse transcribed and 1/10th of the cDNA subjected to PCR. The legend indicates the amount, in IU, of HCV RNA equivalence in the PCR. The 0 indicates a serum negative full process control. NTC is a PCR negative control.

In order to ensure that our strategy was able to detect all common HCV genotypes, we evaluated the ability of our assay to detect a range of HCV genotypes present in the BBI Worldwide performance/genotyping panel. This panel consists of naturally occurring plasma specimens from diverse geographical locations with differing viral loads. The results are given in Figure 2 and clearly show that all the major genotypes present in this panel are detected in bisulphite converted samples using our assay. In addition, genotyping can be performed by sequencing as there is sufficient heterogeneity remaining in the samples post simplification.

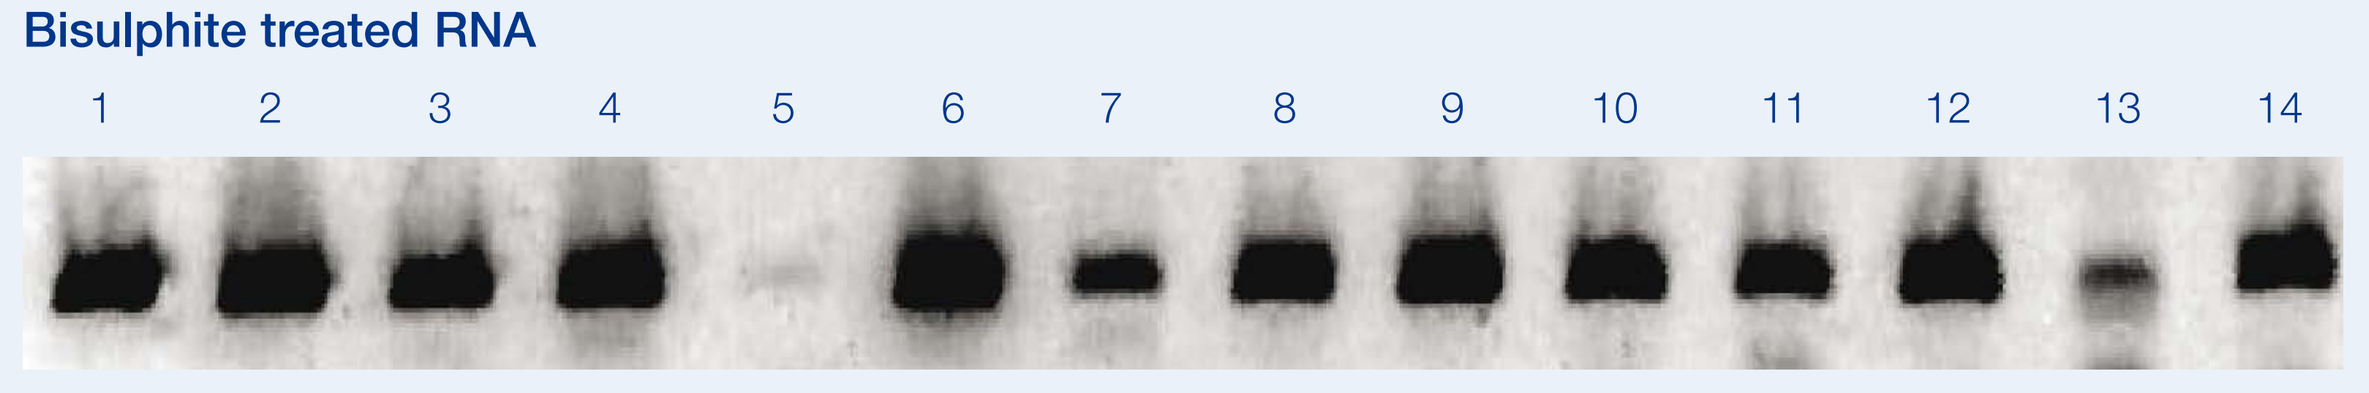
Bisulphite treated RNA
1 2 3 4 5 6 7 8 9 10 11 12 13 14

Detection of HPV by DNA simplification

Sample	1	2	3	4	5	6	7	8	9	10	11	12	13	14
Genotype	1b	1a	1b	2a	2a/c	3b	3a	3a	4a	4	4a	5a	5a	6a
Quantitation Roche COBAS RNA x10^5IU/mL														
	1.3	4.3	1.4	9.1	0.01	24	3.0	7.2	3.8	0.32	1.4	2.4	2.0	6.1
Bayer Versant 3.0 RNA x 10^5IU/mL														
	12	4.3	0.7	5.3	<0.006	17	0.9	4.9	1.7	0.24	0.86	0.84	0.08	1.4

Figure 2: Performance evaluation of HGS HCV assay on a World Wide Performance Panel

Following the demonstrated sensitivity of our assay and the ability to detect various genotypes, we assessed 138 blinded clinical samples and compared the results to those obtained from a range of commercially available HCV assays. A selection of these results are given in Figure 3 and these clearly show that the HGS HCV assay reliably detects HCV RNA in patient samples. Overall, the HGS assay performed well relative to the Bayer Versant HCV RNA 3.0 test and the Roche COBAS AMPLICOR 2.0 test with 132/138 (96%) in agreement. Of the 6 that were not in agreement with one or other of these tests, 3 were positive by our test and had been positive by one of the other tests at an earlier testing but was subsequently assessed to be negative or below the limit of detection of the assay. On repeated testing, these samples were confirmed to be positive by our test and were shown to contain HCV by sequencing. Thus, we believe this illustrates that our test is more sensitive than the other assays. Two of the other three samples tested negative by our test but had been previously shown to be positive by the Roche or Bayer test at least one year prior to our testing. However, very limited amounts of samples were available and so it is likely that we had too little for detection and/or that the samples had deteriorated over the storage period.

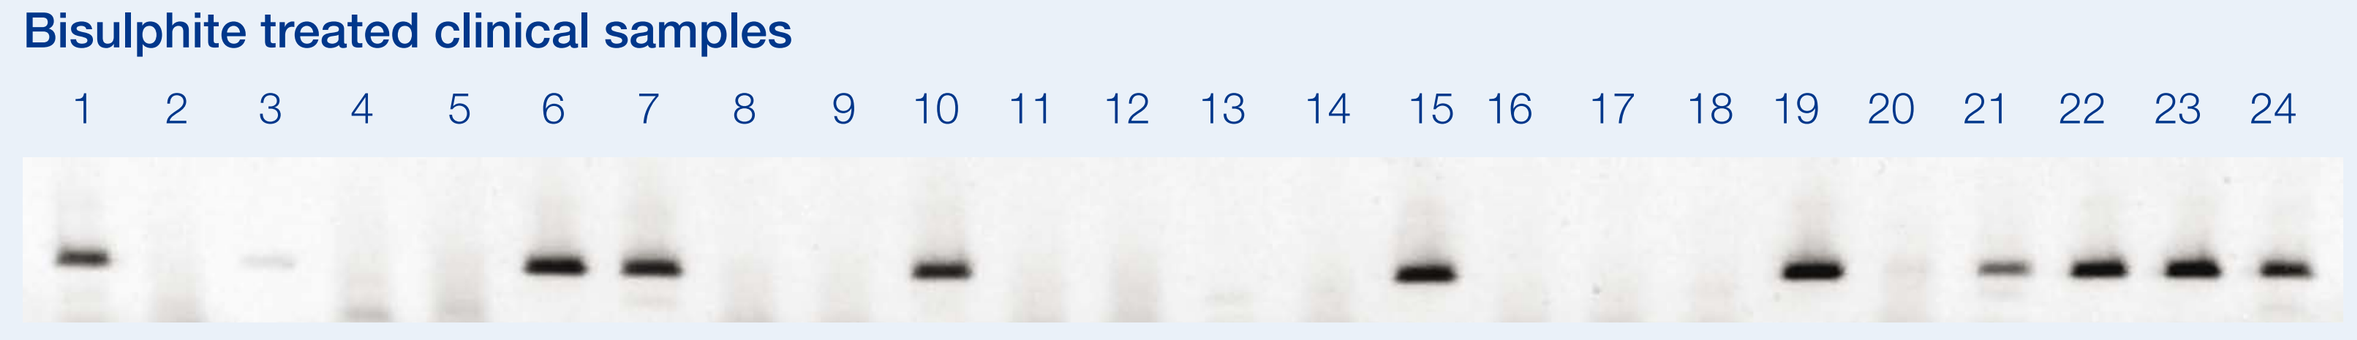
Bisulphite treated clinical samples
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24

Detection of HPV by DNA simplification

Sample	1	2	3	4	5	6	7	8	9	10	11	12
Hospital result	+	-	-	-	-	+	+	-	-	+	-	-
HGS result	+	-	+	-	-	+	+	-	-	+	-	-

Sample	13	14	15	16	17	18	19	20	21	22	23	24
Hospital result	-	-	+	-	+	<615	+	+	+	+	+	+
HGS result	-	-	+	-	-	-	+	+	+	+	+	+

Figure 3: Selection of the results from the blinded clinical samples assayed for HCV RNA by either the Bayer Versant HCV RNA 3.0 or Roche AMPLICOR HCV RNA 2.0 test and the HGS HCV assay.

Assays for other DNA and RNA viruses: Subsequent to the success of our assay in the sensitive and specific detection of HPV and HCV, we assessed our ability to detect a range of other DNA and RNA viruses. Commercially available samples of HBV, CMV, HSV-1, HSV-2, EBV, HIV-1 and EV were obtained and subjected to our simplification technology. A range of concentrations for each virus were purified, simplified and PCR amplified using virus-specific primers. The results of these prototype assays are shown in Figure 4.

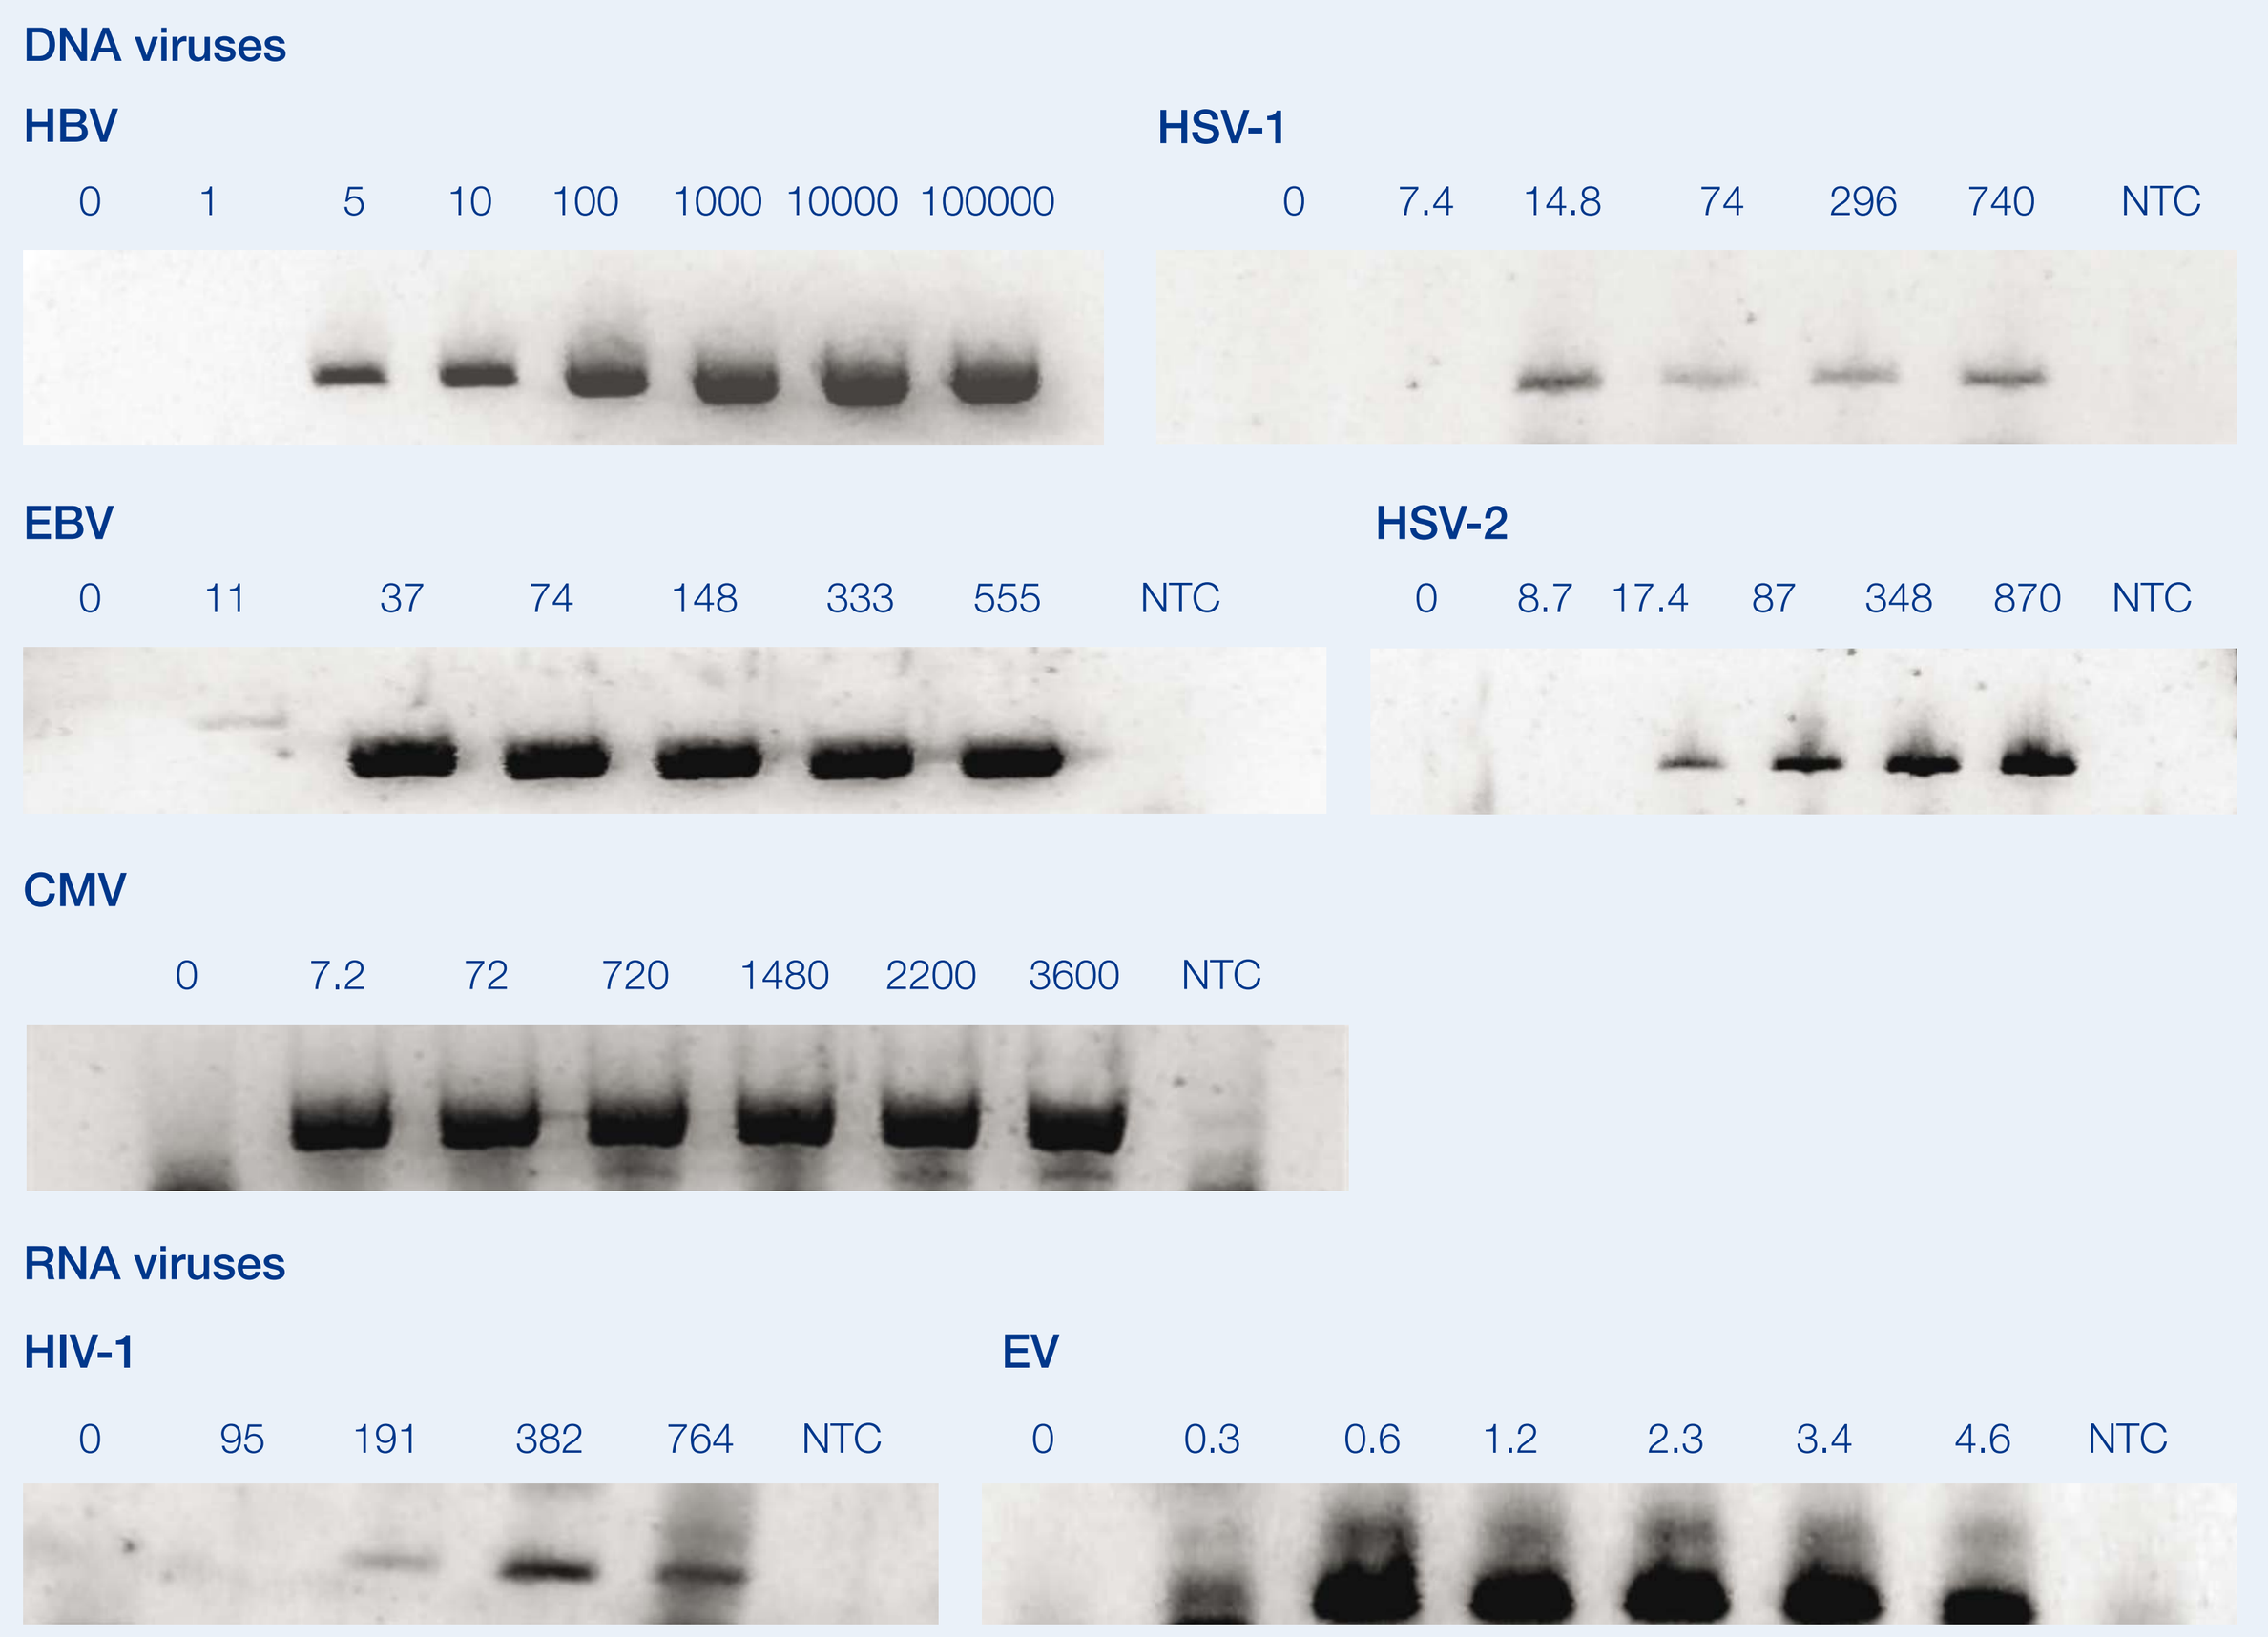
DNA viruses
HBV
0 1 5 10 100 1000 10000 100000
HSV-1
0 7.4 14.8 74 296 740 NTC
EBV
0 11 37 74 148 333 555 NTC
HSV-2
0 8.7 17.4 87 348 870 NTC
CMV
0 7.2 72 720 1480 2200 3600 NTC
RNA viruses
HIV-1
0 95 191 382 764 NTC
EV
0 0.3 0.6 1.2 2.3 3.4 4.6 NTC

Figure 4: Results for prototype assays for DNA (HBV, HSV-1, EBV, HSV-2 and CMV) and RNA (HIV and EV) viruses using the HGS simplification technology. Where known, the concentration of the virus in IU/PCR has been included. For EV, the concentration of virus is given in Enterovirus units (EVU), a proprietary unit that Acrometrix established for their in-house standard, as no recognised international standard exists for EV. NTC = PCR negative control.

As can be seen in Figure 4, a preliminary attempt of our assay to detect other DNA and RNA viruses was successful and we were able to sensitively and specifically amplify a range of medically important viruses, in most cases at concentrations of less than 20 IU/mL sample purified. Further improvements in the sensitivity can be expected with further optimisations. In addition, there is no cross-reactivity between any of the bisulphite primers used above with the other viruses tested (data not shown), which further highlights the specificity of the assay and will allow for multiplexing to detect a range of DNA and RNA viruses in a single sample.

Discussion and conclusions

We have demonstrated that nucleic acid simplification to reduce the 4 base genome to 3 bases is a novel method that enables the detection of multiple viruses. The simplification material still retains sufficient sequence heterogeneity to allow for genotyping or specific species detection. Our technology is adaptable to any nucleic acid detection methods, including isothermal and real-time detection.

Our High Risk HPV detection assay has demonstrated a statistically significant higher Positive Predictive Value ($P<0.001$), lower rate of false positives ($P<0.001$) and greater specificity ($P<0.001$) as compared to the only FDA approved test for HPV detection (the Digene hcII test). This was achieved at a similar level of sensitivity as there was no statistically significant difference between the tests in terms of Negative Predictive Value.

We have developed a novel assay for detecting all strains of HCV based on simplification of the RNA by sodium bisulphite. Bisulphite modification of RNA has not been achieved previously due to the complete degradation of the RNA but our assay has overcome this hurdle. This assay is quick and easy to perform and results in sensitivity over a broad range of HCV concentrations ranging from 1 562 500 IU/mL down to 23 IU HCV/mL. Furthermore, our assay can simultaneously detect all major genotypes of HCV including 1a, 1b, 2a/c, 3a, 3b, 4a, 5a and 6a from a wide range of geographical locations.

The HGS HCV assay was used to assess a blinded panel of 138 clinical samples and was shown to perform well relative to the Bayer Versant HCV RNA 3.0 test and the Roche COBAS AMPLICOR 2.0 test with 132/138 (96%) samples in agreement. Of the six discordant samples, four were positive by the HGS HCV assay, and the presence of HCV in the samples was confirmed by sequencing, indicative of an increased level of sensitivity of our assay in comparison to the other tests.

HGS simplification technology has also been successfully applied to the specific and sensitive detection of a range of other medically important DNA and RNA viruses including HBV, HSV-1, HSV-2, EBV, CMV, HIV-1 and EV. Multiplexing to detect a range of viruses in a single reaction should also be easily achievable and will make the HGS assay a versatile tool in the molecular diagnostics industry.

Human Genetic Signatures

Convert Simplify Understand

Human Genetic Signatures Pty Ltd
ABN: 30 095 913 205

Email: info@geneticsignatures.com
Phone: + 61 2 9870 7580
Fax: + 61 2 9889 4034
Postal Address: PO Box 184 North Ryde
NSW 1670 Australia